EMBO|EMBL Symposium: The Human Microbiome

Developing a New Generation of First-in-class High Diversity Microbiome Biotherapeutics to Treat Life-threatening Diseases

Lilia.Boucinha[1], Marianne.Robin[1], Johanne.Aubin[1], Marianne.Borderes[1,2], Cyrielle.Gasc[1], Carole.Schwintner[1] and Joël.Doré[1,3]

[1]MaaT Pharma, Lyon, France
[2]Université de Lyon, Lyon, France
[3]INRA, Institut National de la Recherche Agronomique, US1367 MetaGenoPolis, 78350, Jouy en Josas, France

INTRODUCTION

- Intensive treatments such as chemotherapy or the use of antibiotics can significantly stress the gut ecosystem. This is believed to have a major impact on patient's clinical outcome, quality of life and response to treatment.
- Reintroducing microbial species in patients has allowed unprecedented progress in the treatment of *C. difficile* infections (CDI).
- We analyzed several studies that correlate high gut microbiota diversity with an improved clinical outcome in various diseases. Thus, to improve microbial diversity and reduce species heterogeneity of the biotherapeutic product manufactured in our GMP platform, we developed a technology based on pooling healthy donor microbiota.
- The first batch-reproducible, high diversity product is currently tested in an interventional MaaT Pharma clinical trial (ClinicalTrials.gov: NCT NCT03359980) for treating steroid refractory gastrointestinal (GI) acute Graft vs Host Disease (GvHD) after allogenic hematopoietic stem cell transplantation (alloHSCT). This trial is ongoing in Europe with the objectives to demonstrate the GI and overall GvHD response as well as a good safety profile of the product in this indication.

Manufacturing Process

Eight healthy donors qualified for our first MaaT013 GMP production campaign, after screening and testing a database of 2 800 volunteers (donor questionnaire, physician consult, quick test on stool, complete blood and stool screening). Qualified donors participated on a daily basis over a period of 5 weeks during which 341 drug products were manufactured.

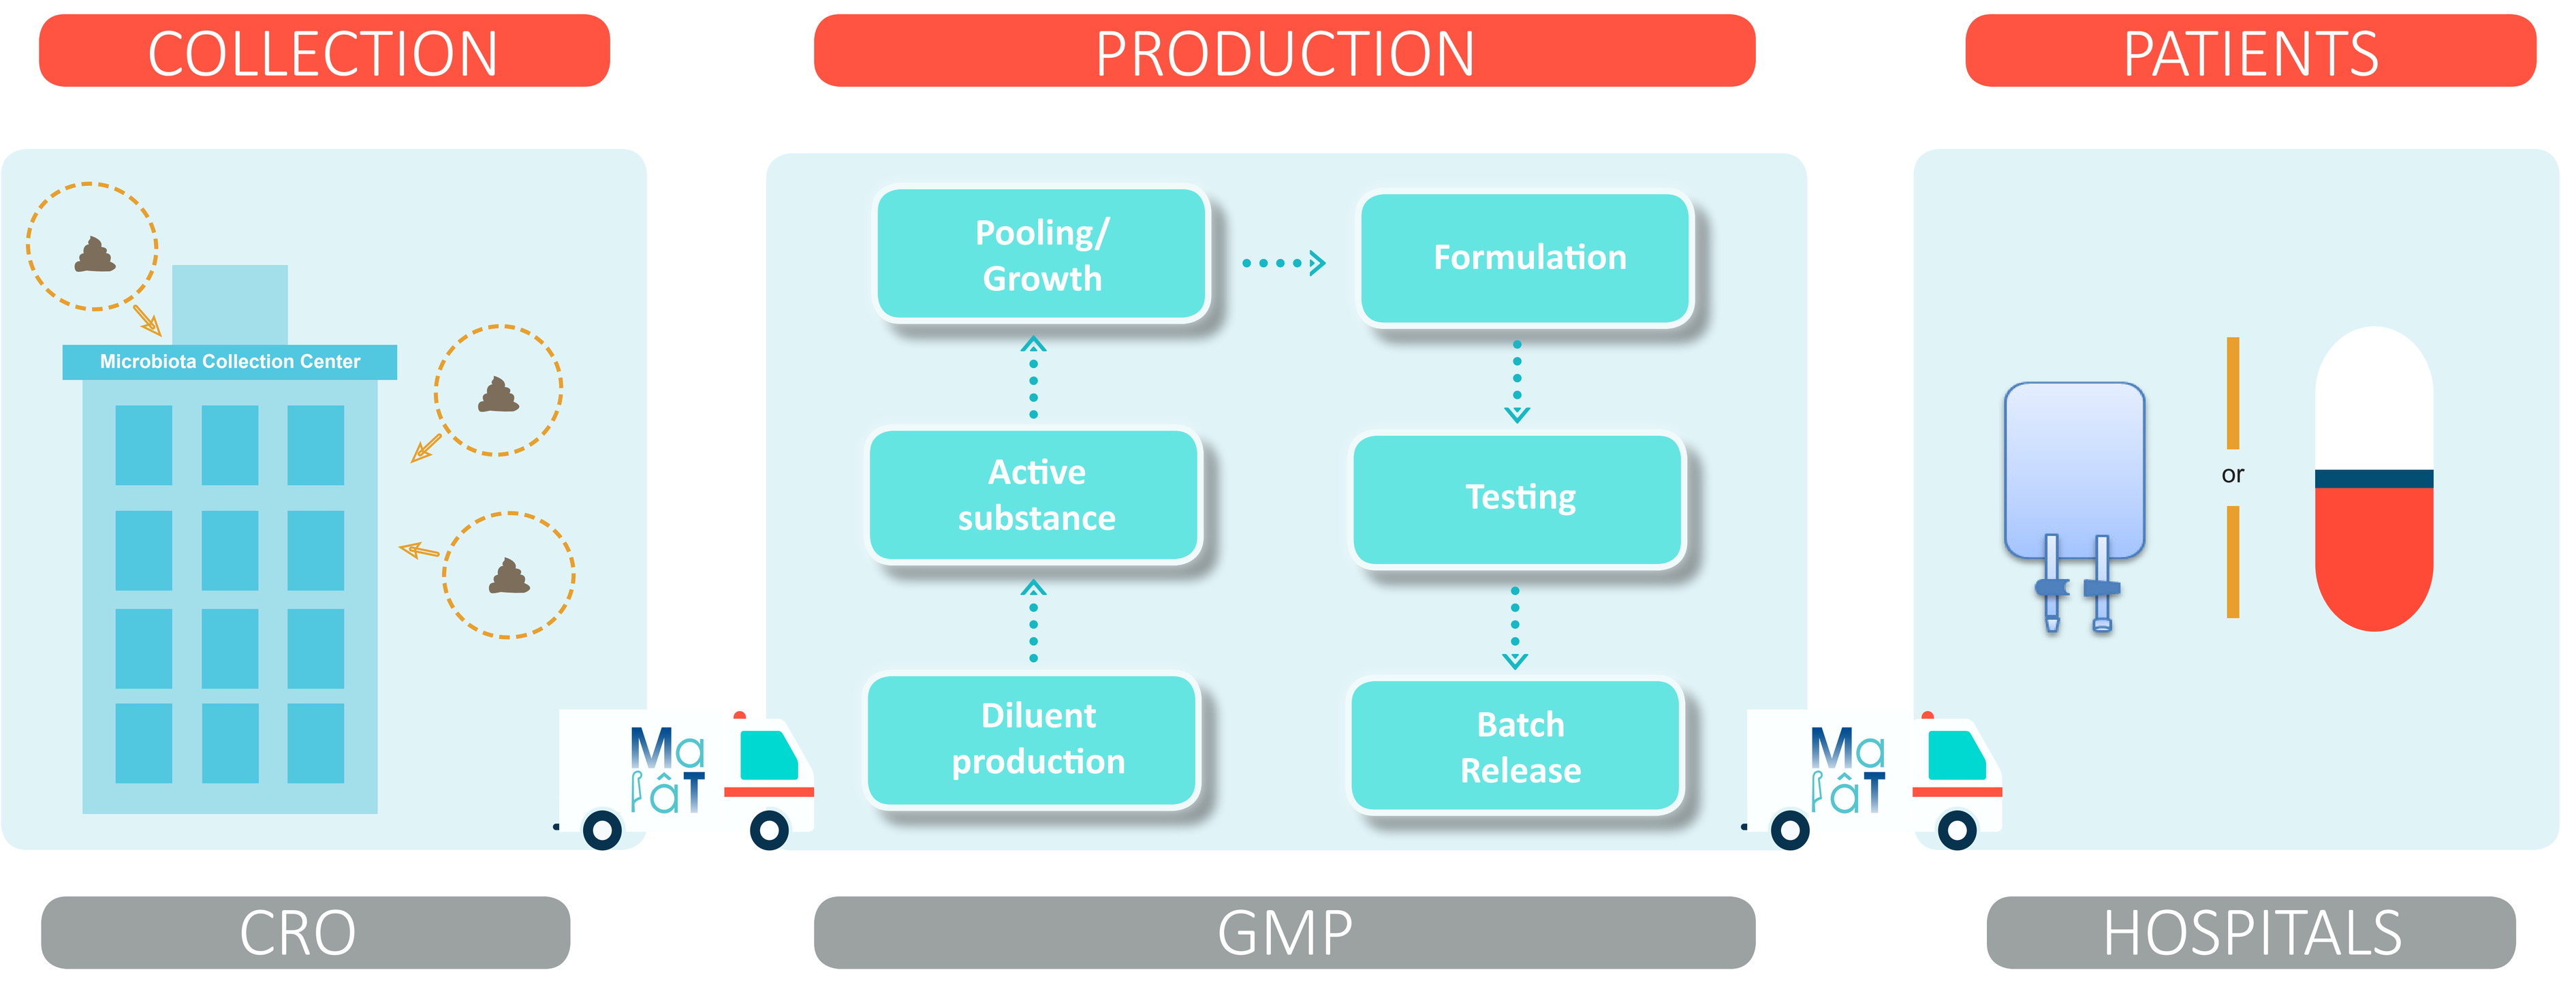
COLLECTION
PRODUCTION
PATIENTS
Microbiota Collection Center
Pooling/ Growth
Formulation
Active substance
Testing
Diluent production
Batch Release
or
CRO
GMP
HOSPITALS

MaaT Pharma biotherapeutic drug manufacturing process

Two quality control criteria for product release were based on 16S analysis results: (1) *Proteobacteria* relative abundance <5% and (2) Inverse Simpson index >4.
A reference sample with known profile was included in the MaaT 16S analysis process. This reference allows standardizing metagenomics analysis and validating all analysis steps, from DNA extraction to bioinformatics analysis.

16S Data Analysis Results

16S analysis (V3-V4) was performed on 59 samples: 24 pools (1 sample/batch) and 8 donors (from 1 to 9 samples/donor). For each sample, a minimum of 140 000 Paired-End reads were generated (2 x 300bp, Illumina MiSeq v3).

De novo OTU and diversity analyses were performed with MaaT Pharma **gʌt Print**® - 16S analysis pipeline. Taxonomy and diversity analyses were realized on rarefied data (60 000 amplicons/sample).
The entire analysis workflow used for product analysis was validated and qualified in order to deliver GMP compliant results.

Taxonomy Analysis

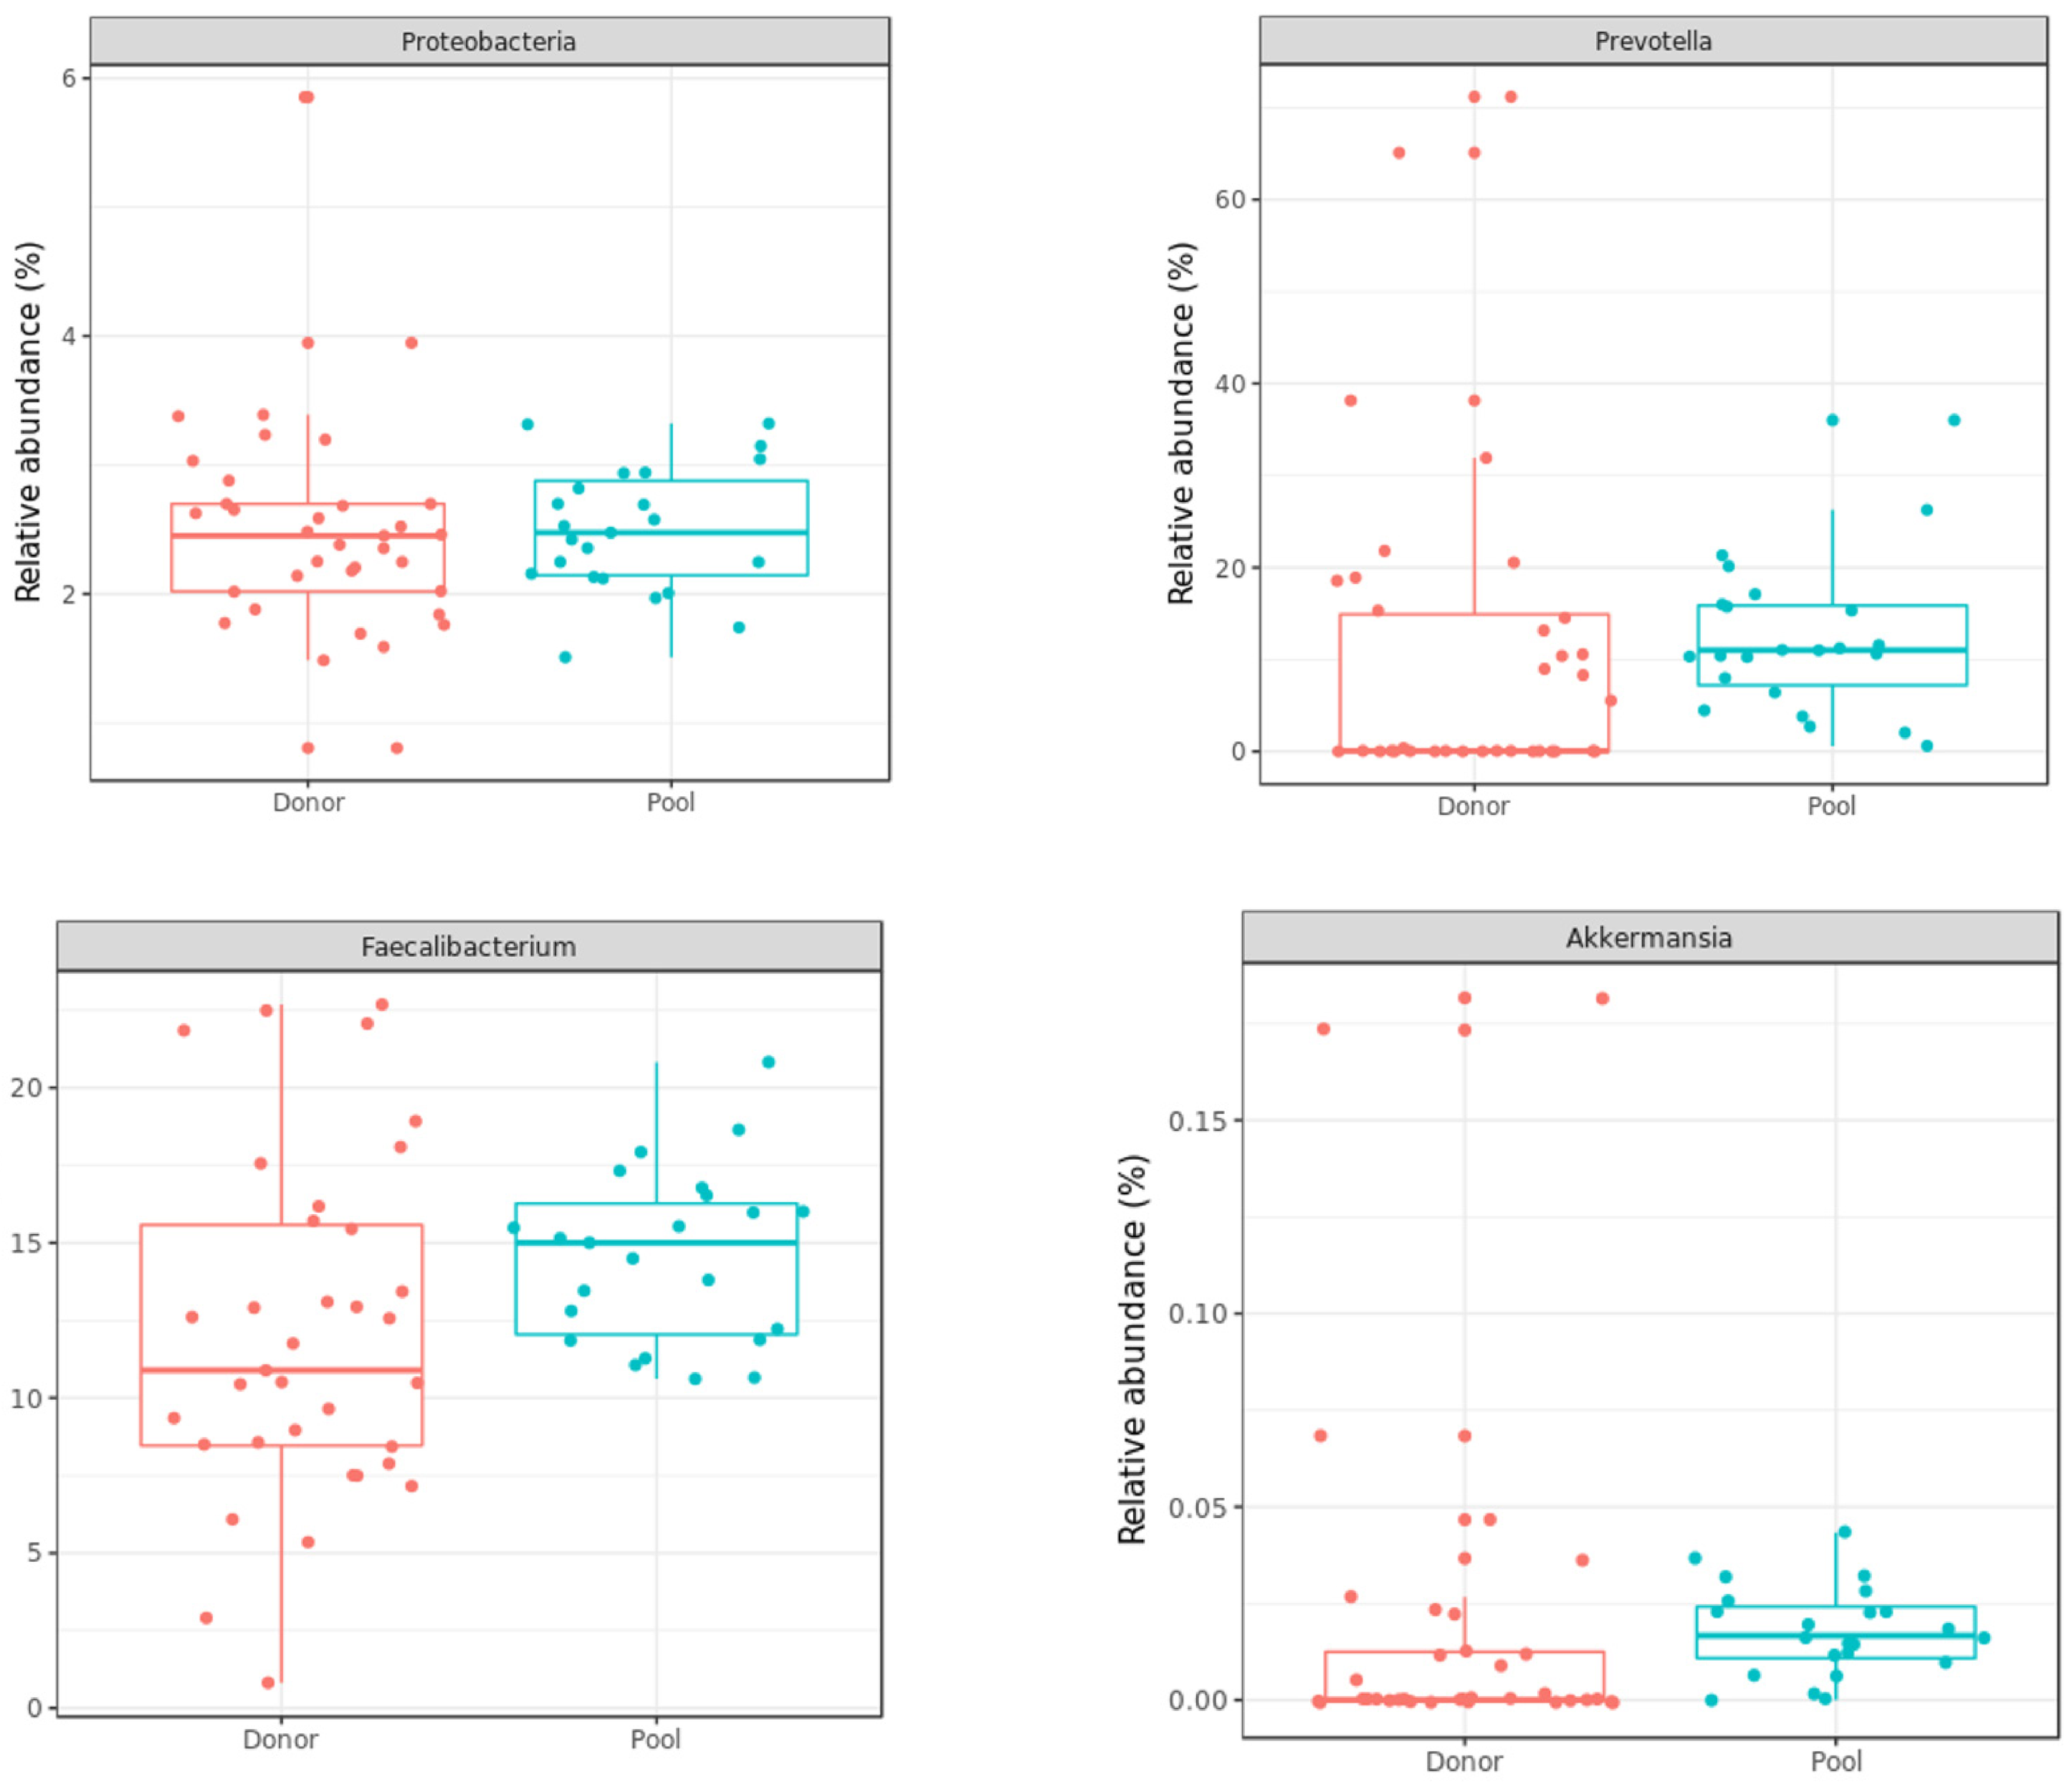
Proteobacteria
Prevotella
Faecalibacterium
Akkermansia
Relative abundance (%)
Donor
Pool

Less than 5% of Proteobacteria is observed in all pooled products.
Increased proportion of prevalent genera (present in more than 90% of samples) from 40% to 76%; among them are found beneficial genera such as *Akkermansia* (present in 50% of donors and 91% of pooled products, becoming therefore prevalent in pools).
For genera such as *Faecalibacterium* or *Prevotella*, the relative abundance is higher and less variable in pooled products.

Alpha-Diversity Analysis

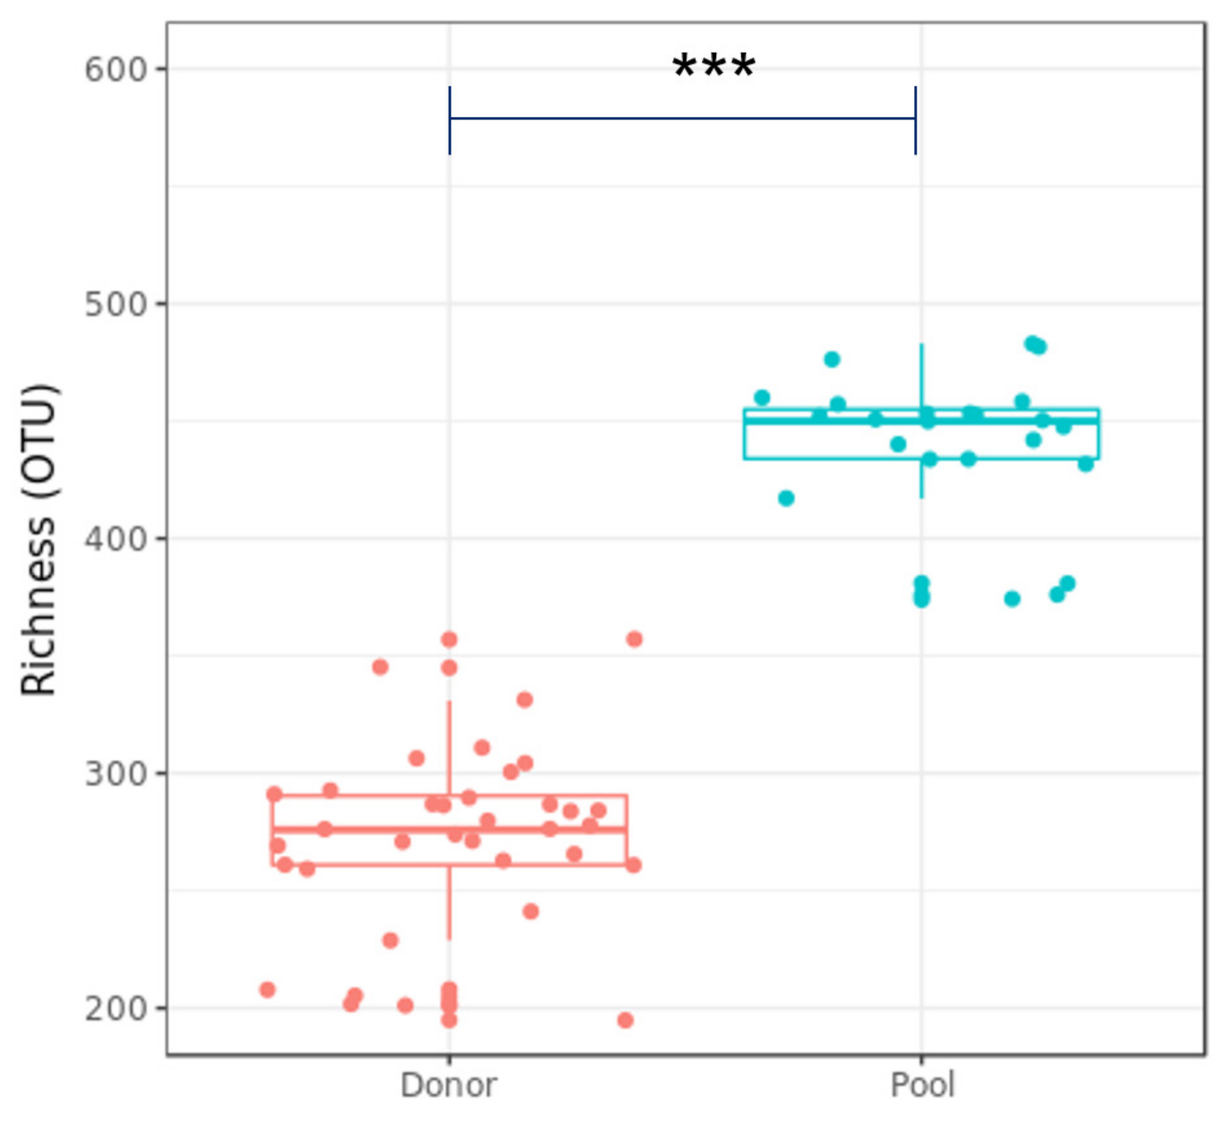
Richness (OTU)

Donor
Pool

Richness increase by 58% in pooled products.

Significant reduction of relative standard deviation.

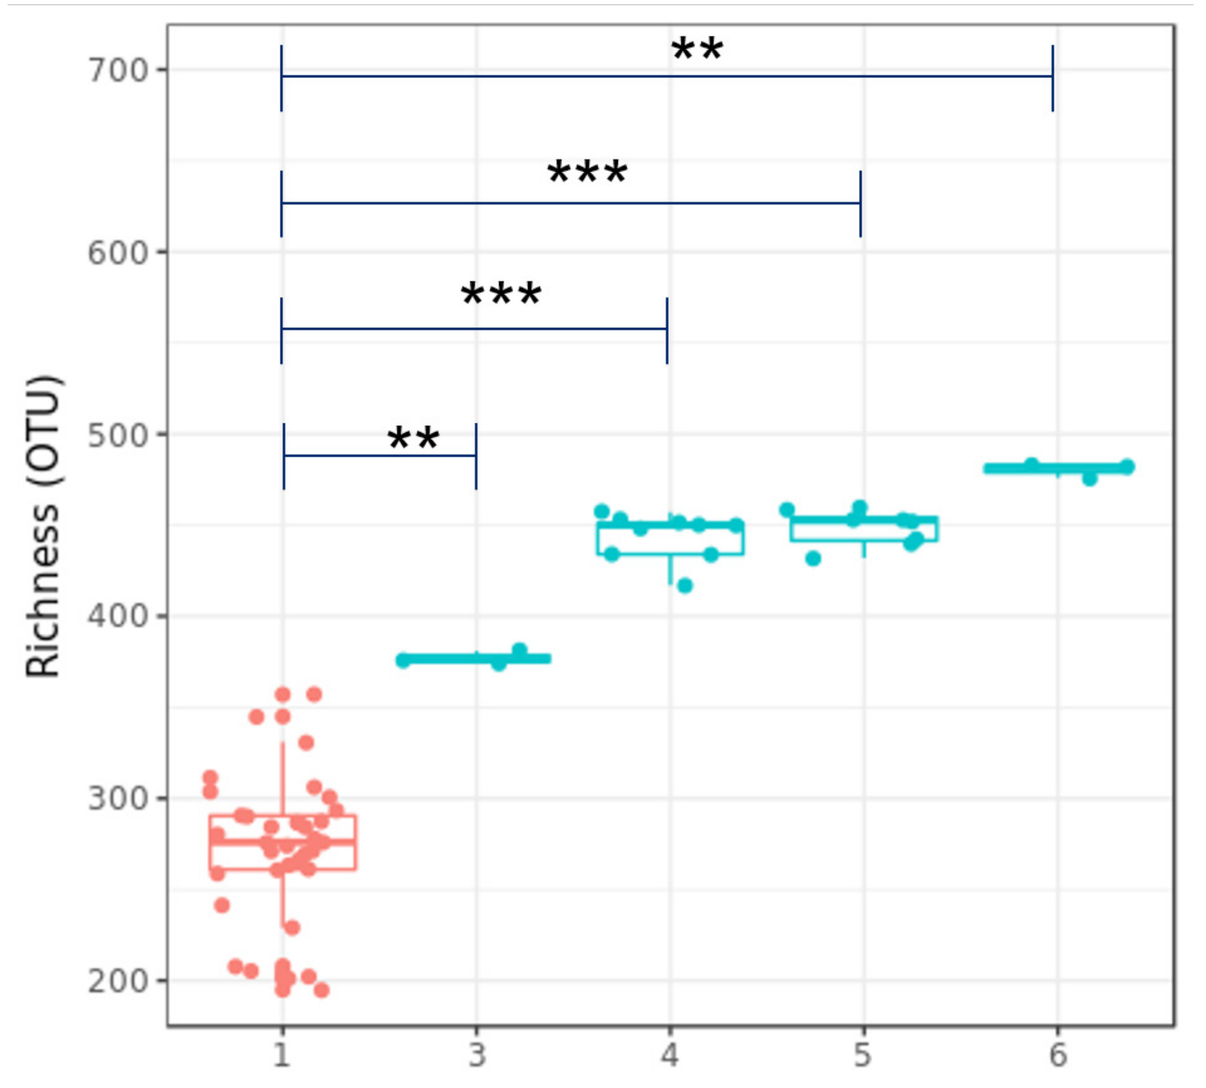
Richness (OTU)
**

**
1
3
4
5
6

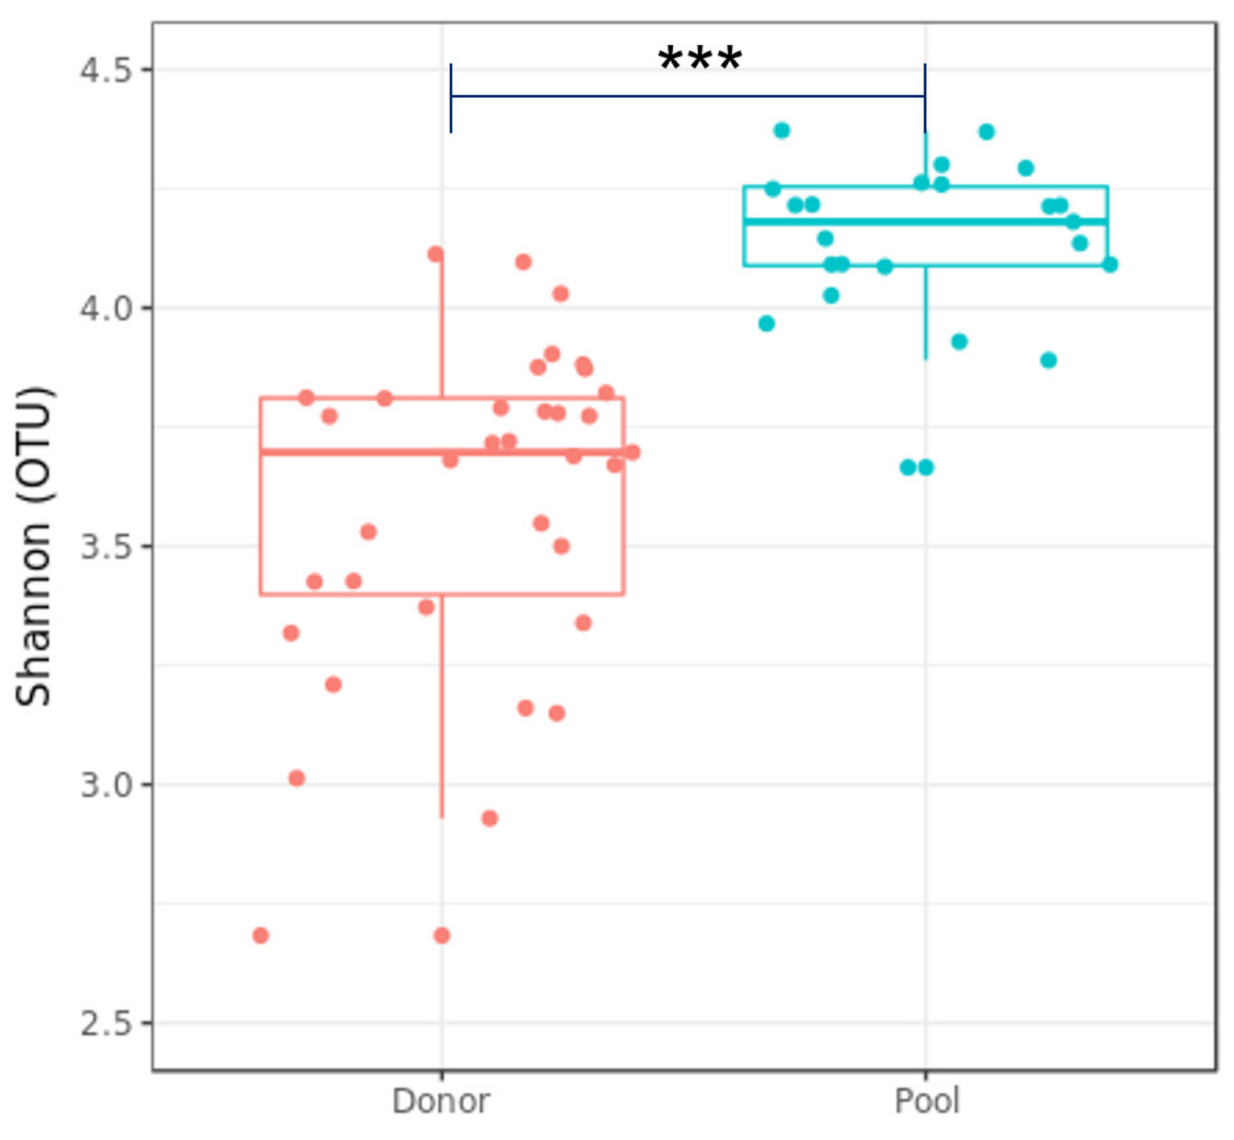
Shannon (OTU)

Donor
Pool

Shannon Diversity increase by 11% (InvSimpson -30%) in pooled products.

Significant reduction of relative standard deviation.

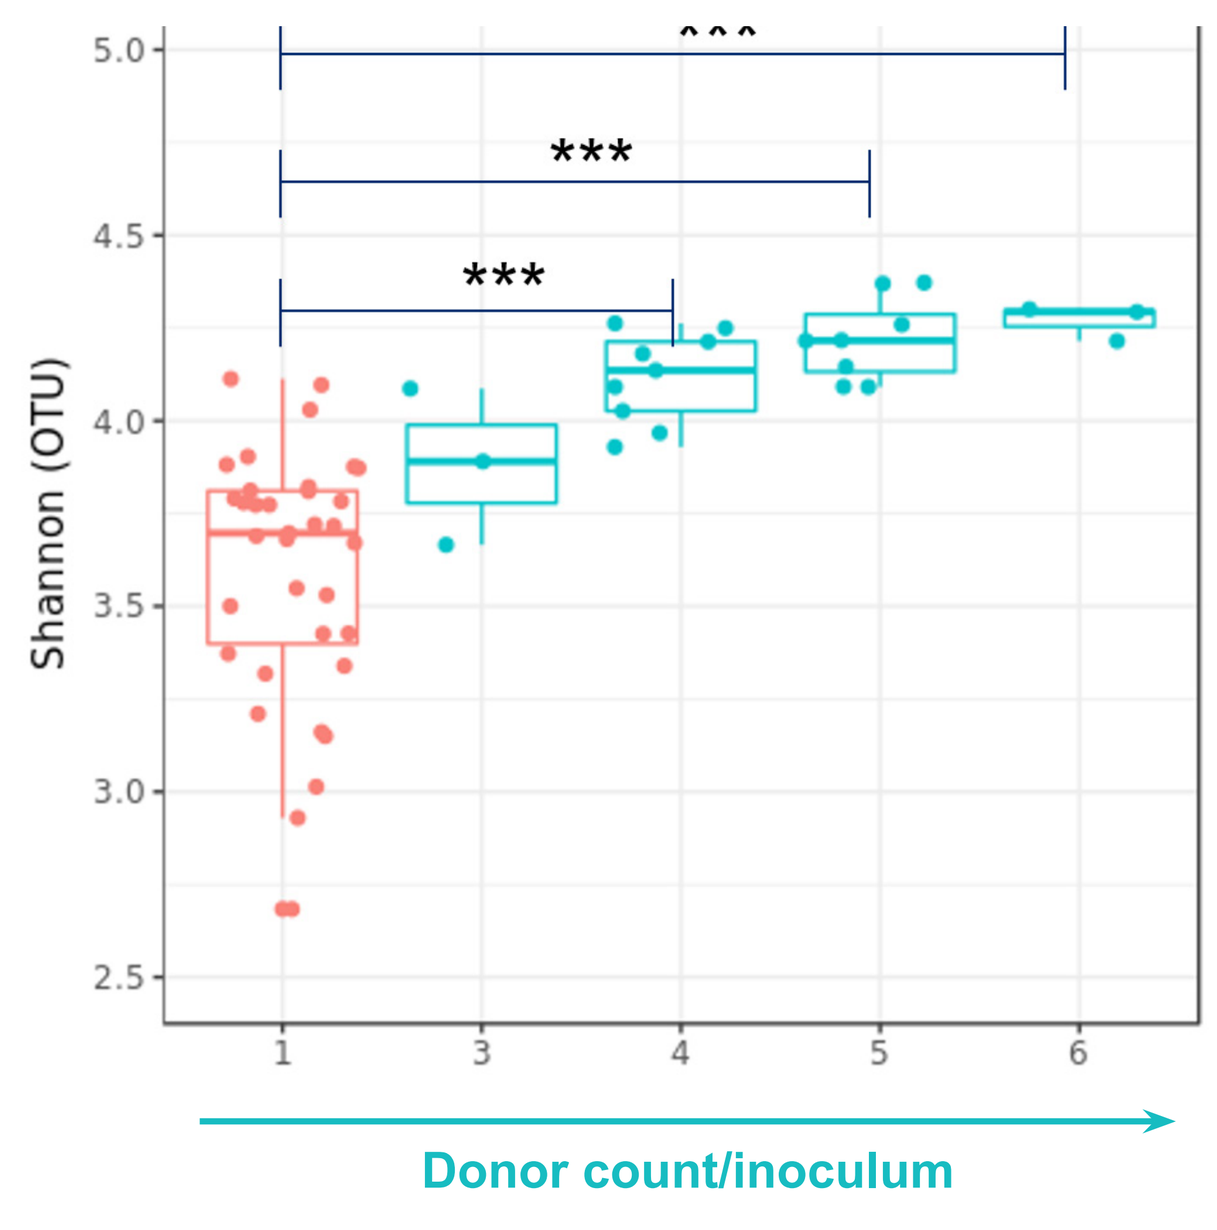
Shannon (OTU)

1
3
4
5
6
Donor count/inoculum

Pooled products are characterized by a **significantly higher richness and diversity**; moreover their **diversity values are significantly less variable.**

Pooled products **richness and diversity are increasing with the number of participating donors are less variable.**

Beta-Diversity Analysis

Principal Coordinate Analysis based on Bray-Curtis dissimilarity

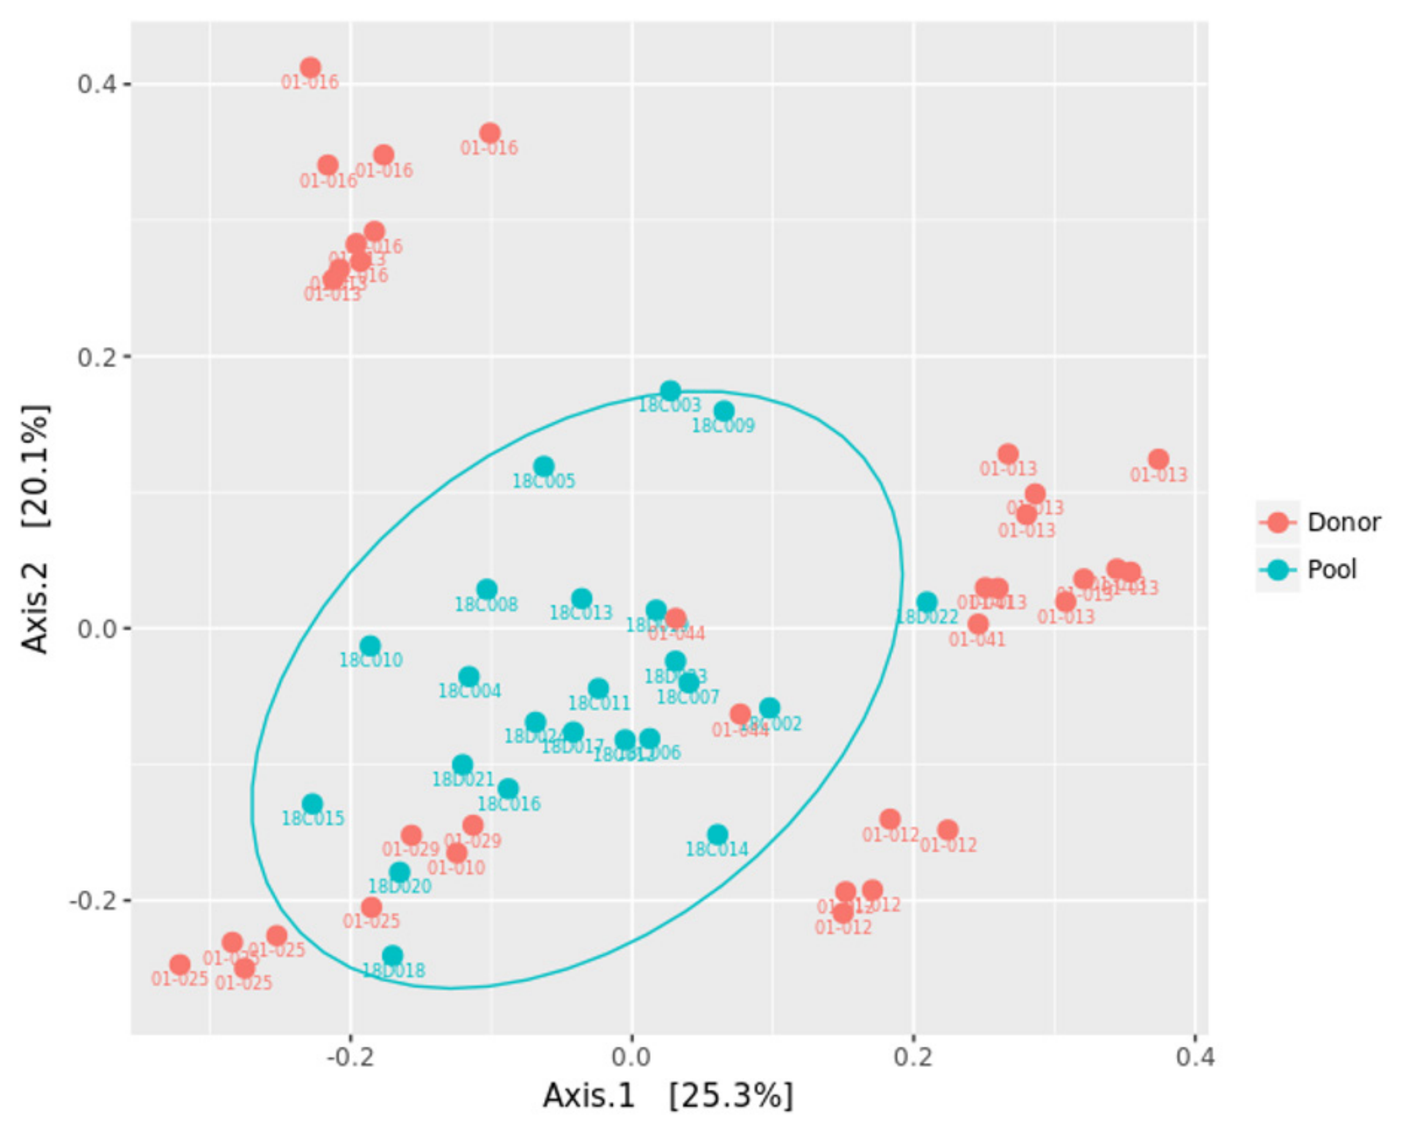
Axis.1 [25.3%]
Axis.2 [20.1%]
Donor
Pool

Pooled product samples are concentrated around the barycenter of donor samples

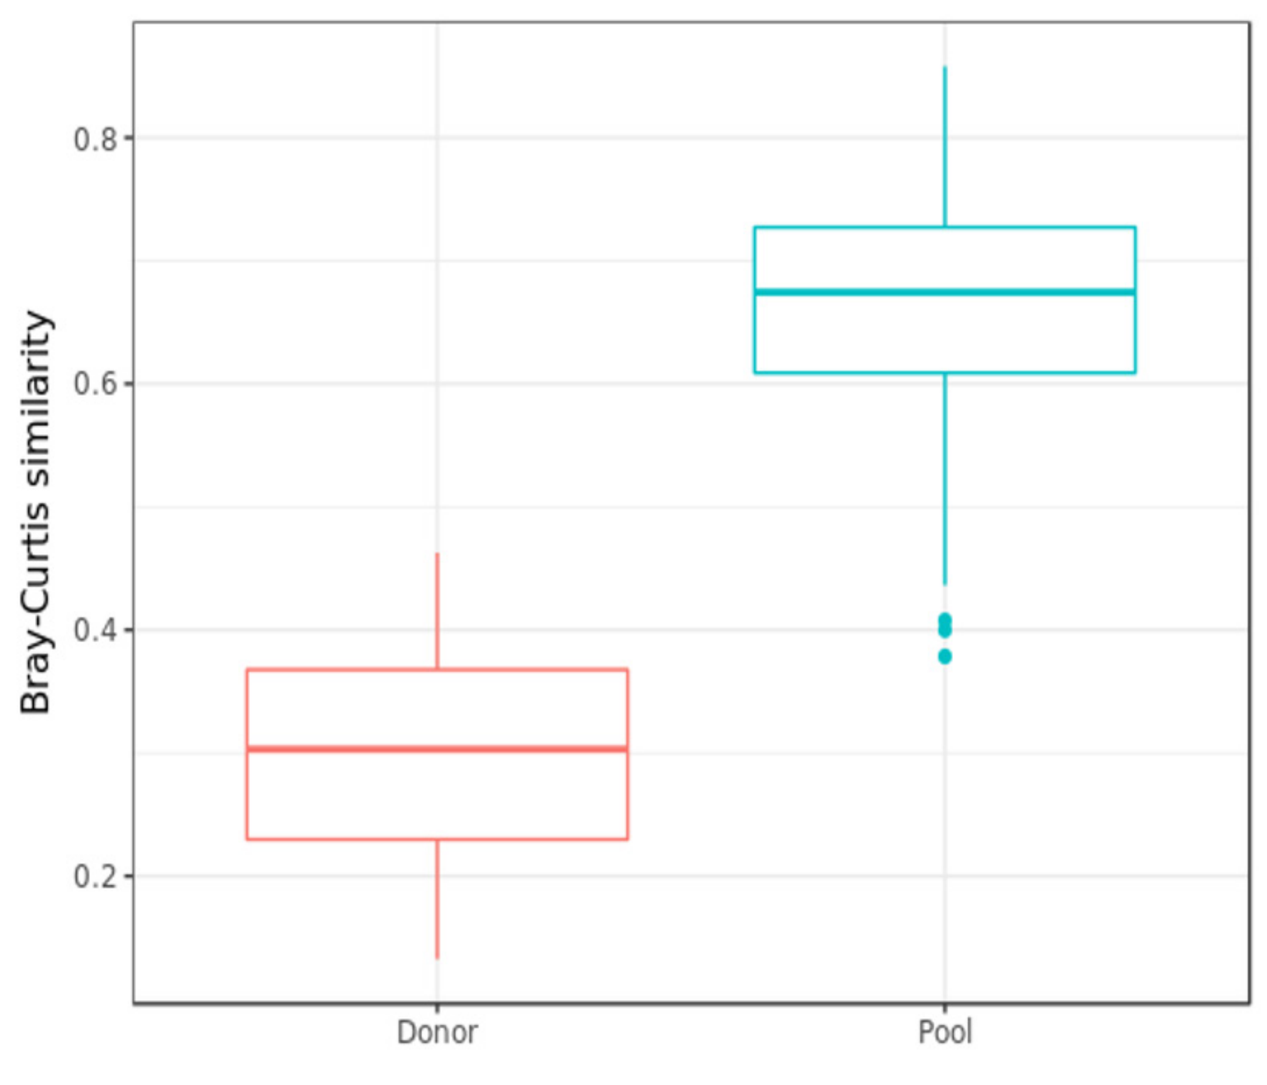
Bray-Curtis similarity
Donor
Pool

Greater homogeneity of pooled products compared to individual donors

Conclusion

MaaT Pharma established a process to develop a standardized, unique and reproducible high microbial diversity biological product compared to other allogenic replacement products: greater homogeneity, decreased variability in alpha-diversity values, increased genus prevalence. Recent studies show that the presence of a taxon in recipient microbiome, prior to patient reintroduction, is increasing the chances of engraftment of most closely related taxa. Therefore, significantly higher richness of our product increases the chances of engraftment of more species than would an individual donor-based product. Our safe differentiated product should overcome donor-dependent responses and improve treatment success. Ongoing shotgun analysis will allow a deeper characterization of pool based drug-products.

@MaaT_Pharma
MaaT Pharma